Biochemical characterization of AB-343, a novel, potent, and orally bioavailable SARS-CoV-2 main protease inhibitor with a pan-coronavirus profile

Kayleigh R. McGovern-Gooch*[1], Nagraj Mani [1], Andrea Cuconati [1], Xiaohe Wang [1], Dimitar Gotchev [1], Kristi Y. Fan [1], Klaus Maskos [2], Chia-Yi Chen [2], Archna Prasad [2], Konstanze von König [2], Debora L. Konz Makino [2], Andrew G. Cole [1], Angela M. Lam [1], Michael J. Sofia [1]

[1] Arbutus Biopharma, Inc., Warminster, PA, USA and [2]Proteros Biostructures GmbH, D-82152 Martinsried, Germany

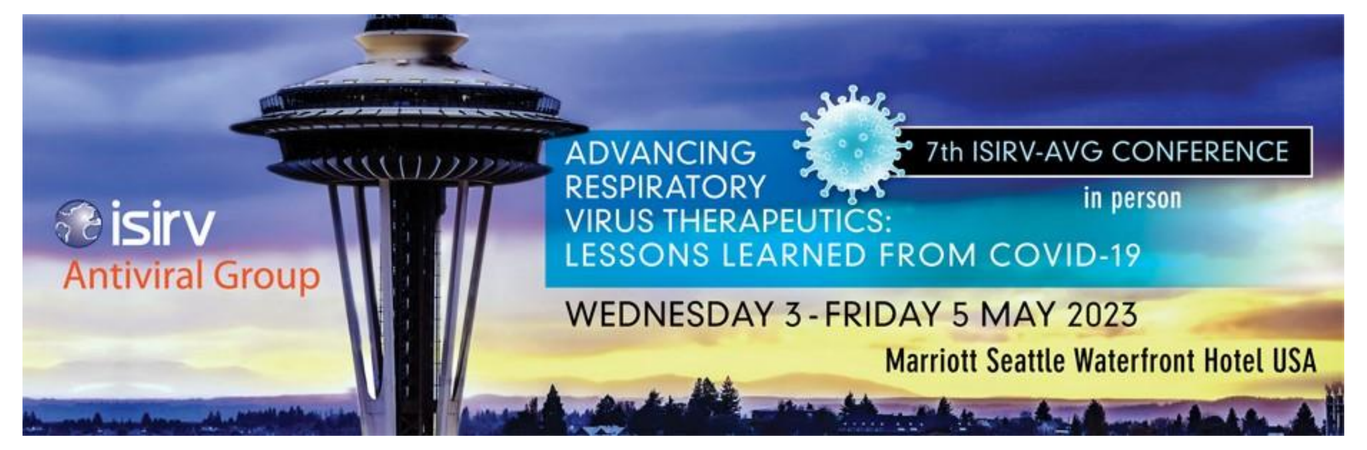
isirv
Antiviral Group
ADVANCING RESPIRATORY VIRUS THERAPEUTICS: LESSONS LEARNED FROM COVID-19
7th ISIRV-AVG CONFERENCE
in person
WEDNESDAY 3 - FRIDAY 5 MAY 2023
Marriott Seattle Waterfront Hotel USA

#3

Arbutus BIOPHARMA

BACKGROUND

- Potent, orally available treatments for COVID-19 are urgently needed.[1]
- Although still protective against severe disease, vaccine efficacy against infection wanes quickly, especially against recent Omicron variants.[2]
- In addition to acute infection, long COVID affects millions of people, with its causes and treatments still unknown.[3]
- SARS-CoV-2 main protease (Mpro) is responsible for processing the viral polypeptide into non-structural proteins and is essential for replication.
- The active site of Mpro is highly conserved among human coronaviruses; therefore, Mpro is an attractive target for the development of pan-coronavirus treatments.

MATERIALS AND METHODS

Mpro enzymes: All Mpro enzymes were expressed and purified from *E. coli.*

Mpro enzyme assays: Cleavage of a fluorogenic peptide substrate by recombinant Mpro was used to determine enzyme activity. Reactions were monitored continuously for 90 minutes at room temperature, and initial slopes were used to calculate fractional velocity for each reaction. K_i values were calculated using the Morrison equation, and k_{cat} and K_M values were calculated using the Michaelis-Menten equation in GraphPad Prism (Boston, MA).

Surface plasmon resonance (SPR) assays: Biotinylated Mpro was immobilized on SA chip. The binding between testing compounds and Mpro were analyzed with single cycle kinetics mode and kinetic parameters were obtained using Biacore Insight Evaluation Software.

Thermal shift analysis: Melting temperature (T_m) of Mpro in the presence or absence of inhibitor was determined by fluorescence monitoring of SYPRO Orange using QuantStudio RT-PCR system (Roche).

RESULTS

AB-343 shows pan-coronavirus activity

- AB-343 potently inhibits SARS-CoV-2 Mpro with a K_i of 3 nM in an enzymatic assay.
- AB-343 also inhibits Mpro from SARS, MERS, OC43, HKU1, NL63, and 229E with K_i values ranging from 5 – 45 nM.
- Coronavirus Mpro is a highly conserved target: SARS-CoV-2 Mpro shares 99% full protein sequence similarity with SARS-CoV Mpro, and 60 – 66% similarity with other human coronaviruses.

Mpro	Coronavirus genus	Nirmatrelvir K_i (nM)	Ensitrelvir K_i (nM)	AB-343 K_i (nM)
SARS-CoV-2	**Beta**	**1 ± 0.3**	**1 ± 0.5**	**3 ± 0.8**
SARS-CoV	Beta	2 ± 0.3	1 ± 0.4	5 ± 3
OC43	Beta	11 ± 5	12 ± 4	19 ± 4
MERS-CoV	Beta	35 ± 12	292 ± 191	45 ± 6
HKU1	Beta	5 ± 2	3 ± 1	6 ± 2
NL63	Alpha	142 ± 64	>5000	39 ± 5
229E	Alpha	55 ± 20	>5000	36 ± 14

Table 1: Inhibition activity of nirmatrelvir, ensitrelvir, and AB-343 against Mpro from SARS-CoV-2 and other human coronaviruses. Nirmatrelvir = PF-07321332, the Mpro inhibitor in Paxlovid™ (Pfizer). Ensitrelvir = S-217622, the active ingredient in Xocova® (Shionogi). All data on nirmatrelvir and ensitrelvir shown were generated by Arbutus.

	SARS-CoV-2	SARS-CoV	OC43	MERS	HKU1	NL63	229E
SARS-CoV-2		96.1	48.5	50.3	49.7	44.8	41.1
SARS-CoV	96.1		48.8	51.3	49.0	43.8	40.4
OC43	48.0	48.4		52.9	82.3	43.1	44.0
MERS	50.3	51.3	53.5		55.3	48.5	49.3
HKU1	48.7	48.0	81.5	54.2		43.1	44.7
NL63	43.8	42.8	42.6	47.4	43.0		68.5
229E	40.5	39.9	43.9	48.7	45.0	69.2	

	SARS-CoV-2	SARS-CoV	OC43	MERS	HKU1	NL63	229E
SARS-CoV-2		98.7	65.3	66.3	64.0	62.9	59.6
SARS-CoV	98.7		64.7	67.0	63.3	62.2	59.3
OC43	64.7	64.1		68.6	92.7	63.2	63.9
MERS	66.3	67.0	69.3		68.7	65.2	65.6
HKU1	62.7	62.1	91.7	67.3		63.5	62.9
NL63	61.4	60.8	62.4	63.7	63.3		82.5
229E	58.8	58.5	63.7	64.7	63.3	83.3	

Figure 1: Protein identity (left) and similarity (right) matrices of Mpro from human coronaviruses. Color shading ranges from high identity/similarity in blue to low identity/similarity in red.

RESULTS

Target engagement by AB-343

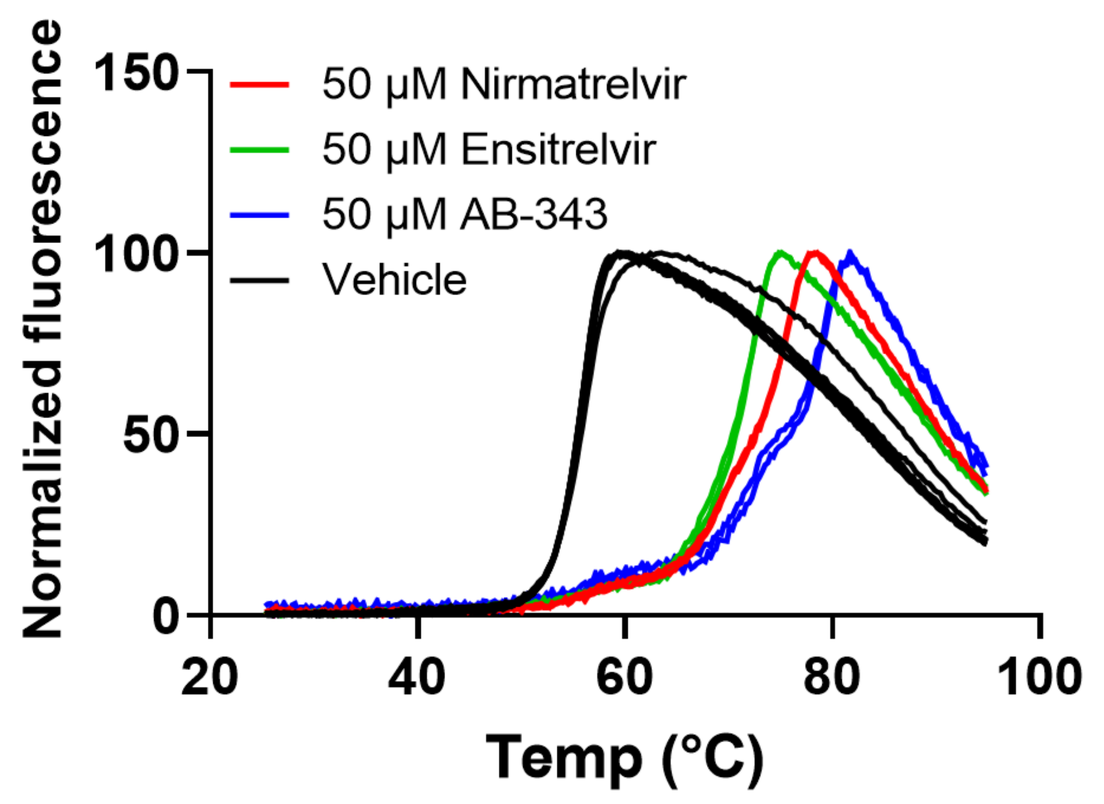
150
100
50
0
20
40
60
80
100
Normalized fluorescence
Temp (°C)
50 µM Nirmatrelvir
50 µM Ensitrelvir
50 µM AB-343
Vehicle

Figure 2. Melt curves of Mpro in presence and absence of inhibitors.

- Binding of AB-343 increases the T_m of Mpro by 23.2 °C, confirming target engagement.
- AB-343 displays slower dissociation measured by SPR, as compared to nirmatrelvir and ensitrelvir.
- Onset of inhibition is rapid, as increasing the preincubation time of AB-343 with Mpro between 5 min and 8 h does not change the IC_{50}.

	Nirmatrelvir	Ensitrelvir	AB-343
ΔT_m (°C)	19.7	16.7	23.2
SPR residence time (min)	21.5	1.1	>99
IC_{50} (nM) Preincubation time: 5 min \| 8 hr	12.3 \| 15.0	8.0 \| 9.2	11.3 \| 11.9

Table 2. Binding and inhibition parameters of Mpro inhibitors.

AB-343 potently inhibits Mpro from prevalent SARS-CoV-2 lineages

- No significant increase in AB-343 K_i is observed against these variants relative to WT in an enzymatic assay.
- P132H, found in Omicron variants, represents 46% of 13.9 million sequences in the GISAID database as of November 21, 2022.[4]

SARS-CoV-2 Mpro Variant	Representative Lineage	Nirmatrelvir K_i fold increase	Ensitrelvir K_i fold increase	AB-343 K_i fold increase
WT (original)	Original	1.0	1.0	1.0
G15S	C.37 Lambda	1.5	1.2	1.2
T21I	B.1.1.318	1.0	0.6	0.7
T24A	B.1.524	1.0	1.5	0.8
L89F	B.1.2	1.0	2.0	0.6
K90R	B.1.351 Beta	1.0	0.9	0.9
P96L	B.1.1.419	1.4	1.0	0.9
P96S	B.1.1.389	1.2	0.7	1.0
P108S	N. 10	1.2	1.0	0.7
P132H	Omicron	1.3	1.3	0.9
A193V	B.1.351.2 Beta	1.0	0.7	0.8
A193T	-	1.0	0.8	0.9
A194S	B.1.617.3	3.5	2.2	2.4
L205V	P.2 Zeta	0.9	0.8	0.7

Table 3: K_i fold increases (K_i against variant Mpro/K_i against WT Mpro) of inhibitors against Mpro from prevalent SARS-CoV-2 variants.

Locations of residues mutated in Mpro variants in relation to AB-343 binding site

- Mpro residues mutated in prevalent SARS-CoV-2 lineages (left) are distal to the active site in which AB-343 (shown in purple) is bound.
- Mpro residues mutated in variants isolated from cell culture and conferring resistance to Mpro inhibitors *in vitro* (right) are closer to the AB-343 binding site.

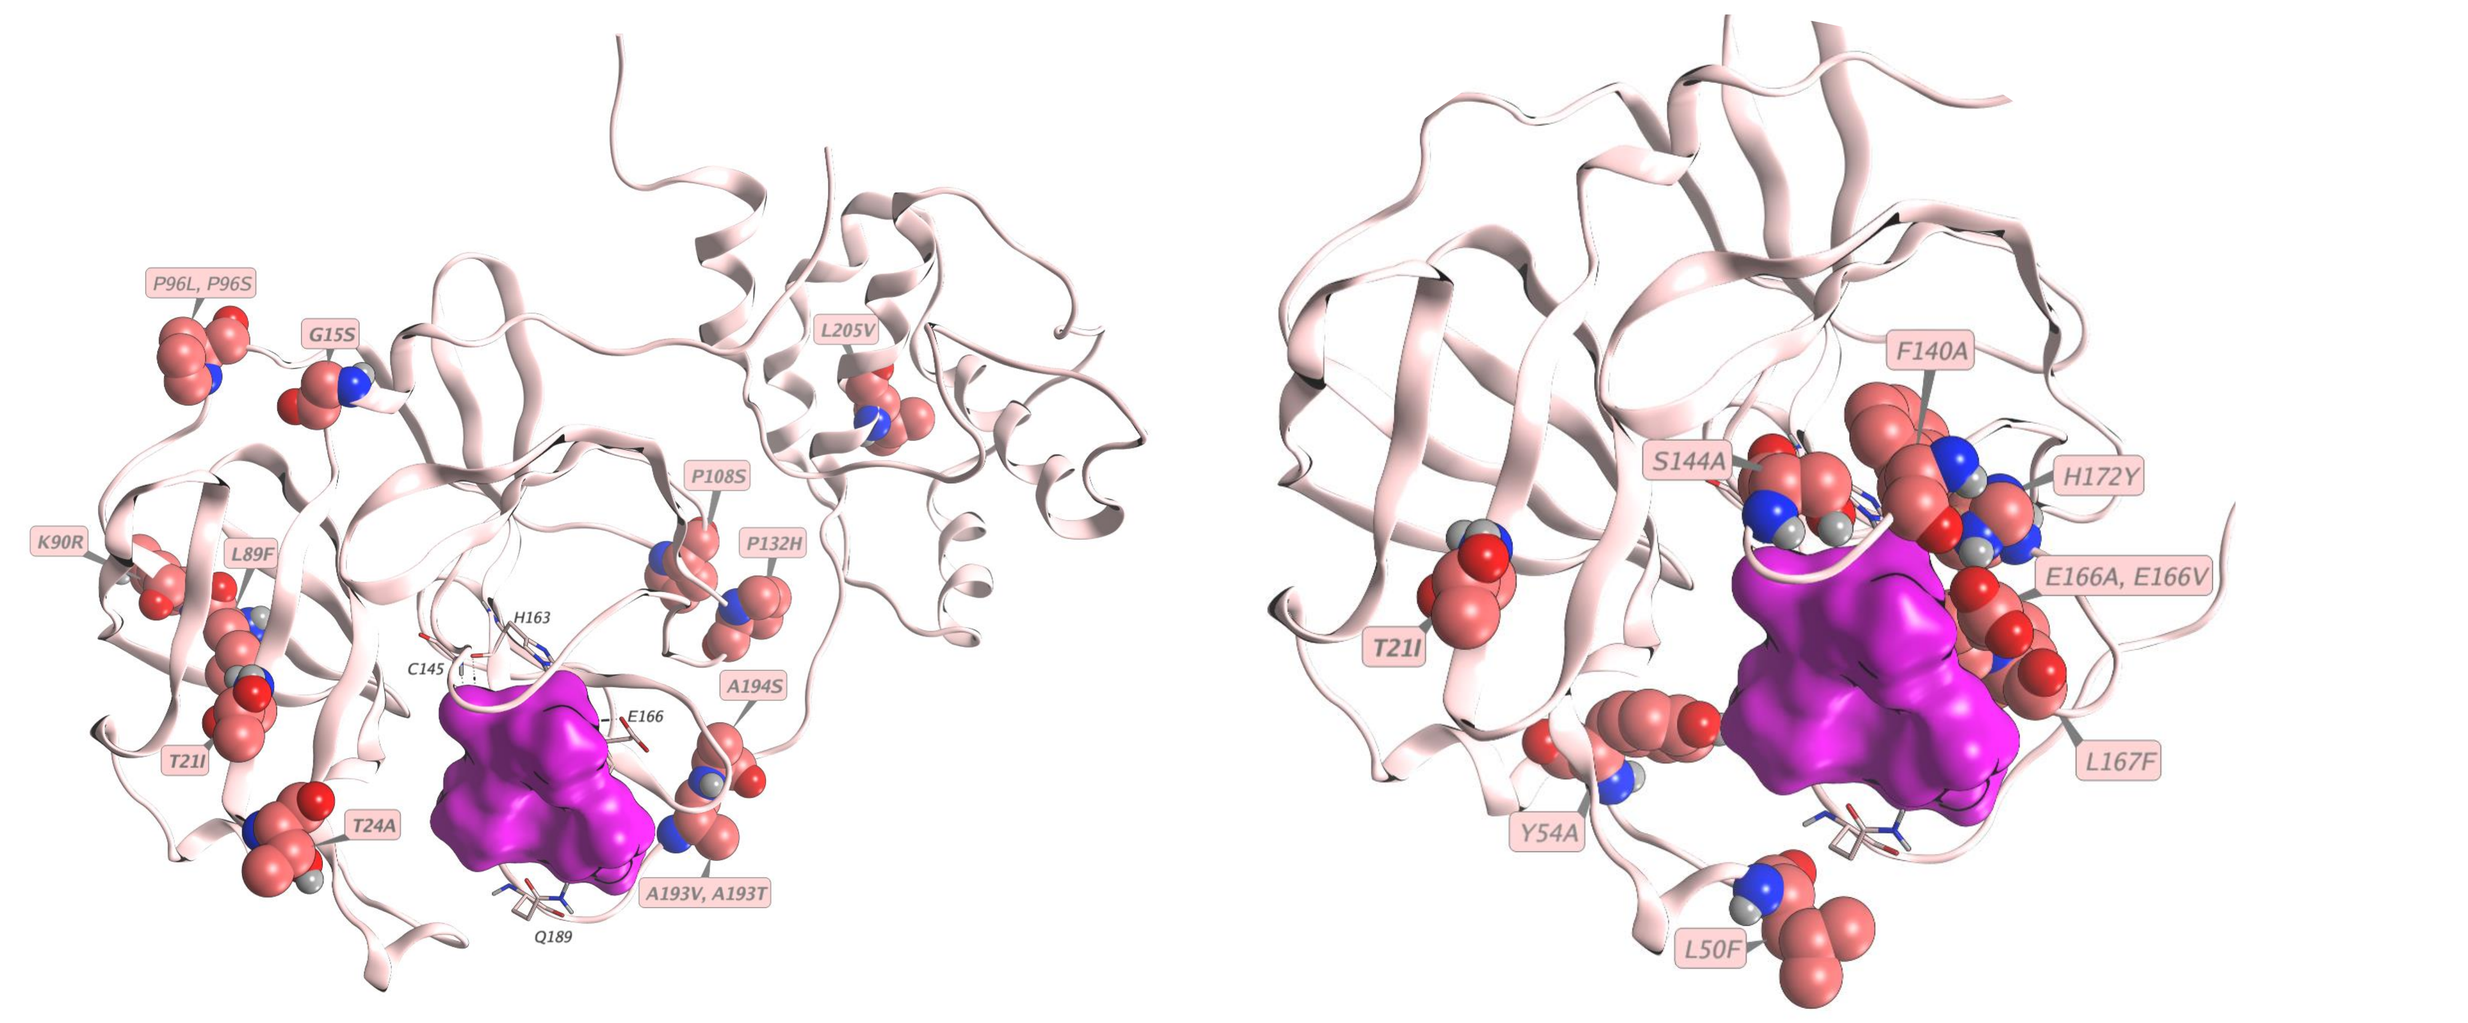
P96L, P96S
G15S
L205V
P108S
P132H
K90R
L89F
H163
C145
E166
A194S
T21I
T24A
A193V, A193T
Q189
F140A
S144A
H172Y
E166A, E166V
T21I
L167F
Y54A
L50F

Figure 3: Structure of Mpro bound to AB-343.

AB-343 exhibits a favorable resistance profile

- Mpro variants were reported to confer resistance to other Mpro inhibitors in cell culture.[5-6]
- AB-343 K_i fold changes against Mpro variants Y54A, F140A, S144A, L167F, and H172Y are lower compared to nirmatrelvir and ensitrelvir.
- Variants containing the E166A/V mutations exhibit cross resistance to AB-343, nirmatrelvir, and ensitrelvir.
- Variants containing the E166A/V mutations are rare: E166A and E166V variants are each found in 0.0001% of all sequences in GISAID.[4]

SARS-CoV-2 Mpro Variant	Nirmatrelvir K_i fold increase	Ensitrelvir K_i fold increase	AB-343 K_i fold increase
Y54A	28.8	89.2	2.2
F140A	41.7	68.1	3.6
S144A	26.0	64.7	2.3
H164N	5.8	11.6	5.0
E166A	90.9	104.8	40.7
E166V	>14203	600.2	>3597
L167F	27.7	25.7	2.6
H172Y	123.8	176.6	5.4
L50F	1.0	0.3	0.7
L50F + E166V	>14203	204.1	2333
L50F + E166V + L167F	>14203	1366	>3597

Table 4: K_i fold increases of inhibitors against Mpro resistance variants.

Catalytic efficiency (k_{cat}/K_M) of Mpro variants is inversely proportional to susceptibility to inhibition by AB-343

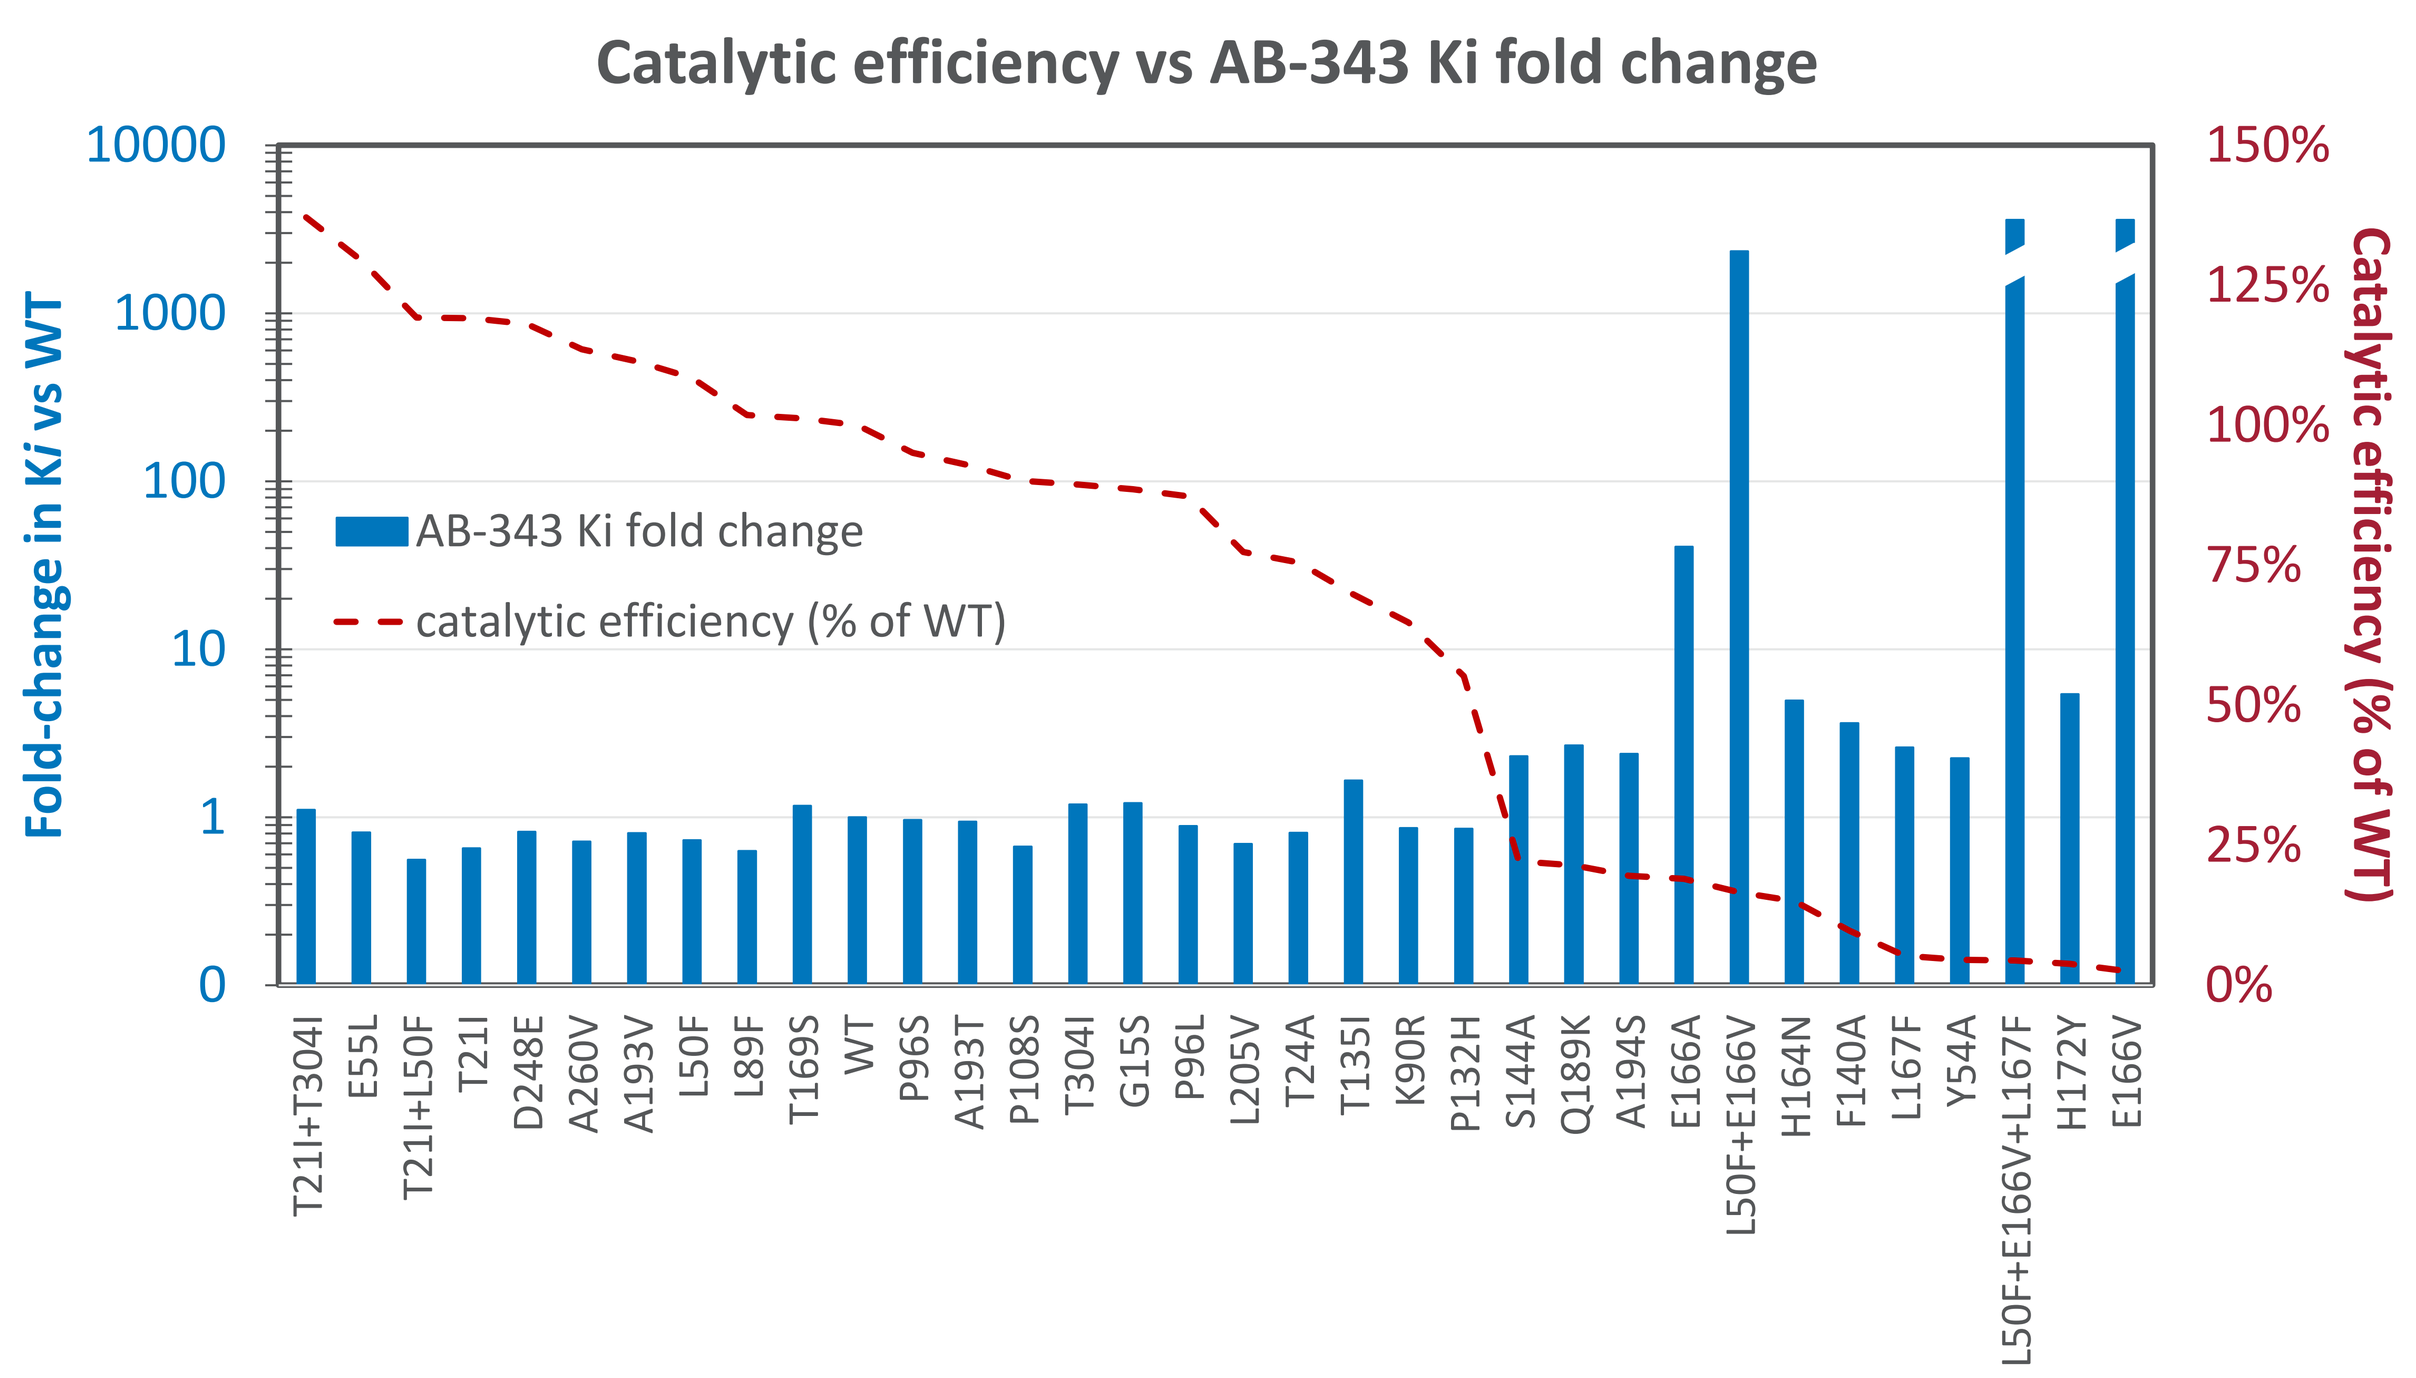
Catalytic efficiency vs AB-343 Ki fold change
10000
1000
100
10
1
0
Fold-change in Ki vs WT
150%
125%
100%
75%
50%
25%
0%
Catalytic efficiency (% of WT)
AB-343 Ki fold change
catalytic efficiency (% of WT)
T21I+T304I
E55L
T21I+L50F
T21I
D248E
A260V
A193V
L50F
L89F
T169S
WT
P96S
A193T
P108S
T304I
G15S
P96L
L205V
T24A
T135I
K90R
P132H
S144A
Q189K
A194S
E166A
L50F+E166V
H164N
F140A
L167F
Y54A
L50F+E166V+L167F
H172Y
E166V

Figure 4: Fold change in AB-343 K_i and catalytic efficiencies for each Mpro variant.

Most Mpro variants with lower catalytic efficiency have reduced thermal stabilities

- P132H, found in Omicron variants, destabilizes Mpro by 1.8 °C relative to WT.
- Variants Y54A, F140A, H172Y, and L50F+E166V+L167F which have the greatest reductions in T_m also have significantly reduced catalytic efficiency (k_{cat}/K_M).

SARS-CoV-2 Mpro Variant	T_m (°C)	ΔT_m (°C)	K_{cat}/K_M (% of WT)
WT	55.8	0.0	100%
P132H	54.1	-1.7	55%
Y54A	52.0	-3.9	5%
F140A	51.1	-4.7	10%
S144A	56.8	1.0	22%
H164N	53.8	-2.0	15%
E166A	53.4	-2.5	19%

SARS-CoV-2 Mpro Variant	T_m (°C)	ΔT_m (°C)	K_{cat}/K_M (% of WT)
E166V	54.1	-1.7	3%
L167F	53.7	-2.2	5%
H172Y	51.7	-4.1	4%
L50F	55.5	-0.3	108%
L50F+E166V	53.5	-2.3	17%
L50F+E166V+L167F	51.1	-4.7	4%

Table 5: Melting temperatures and catalytic efficiencies of Mpro variants. ΔT_m values are relative to WT Mpro T_m.

RESULTS

Mutations conferring resistance reduce thermal stabilization in presence of inhibitors

SARS-CoV-2 Mpro Variant	50 µM Nirmatrelvir ΔT_m (°C)	50 µM Ensitrelvir ΔT_m (°C)	50 µM AB-343 ΔT_m (°C)
WT	19.7	16.7	23.2
P132H	21.0	17.4	24.1
Y54A	13.7	8.5	20.4
F140A	14.1	9.6	20.8
S144A	13.9	6.8	21.4
H164N	19.7	15.2	22.1
E166A	10.0	5.6	13.0
E166V	1.1	4.9	2.4
L167F	17.6	11.2	21.6
H172Y	9.4	4.9	14.3
L50F	20.6	17.9	23.7
L50F+E166V	1.5	6.0	2.9
L50F+E166V+L167F	0.6	2.1	1.8

Table 6: ΔT_m upon inhibitor binding to Mpro variants. ΔT_m values are relative to vehicle T_m of each variant

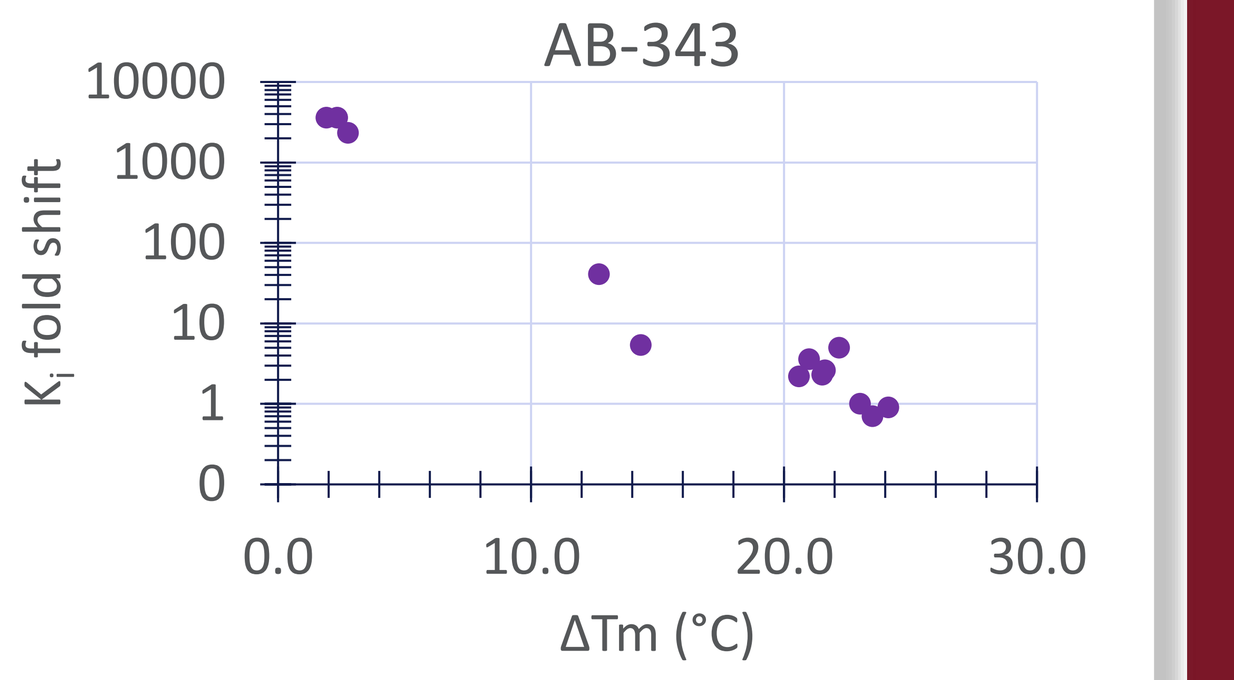
AB-343
10000
1000
100
10
1
0
Ki fold shift
0.0
10.0
20.0
30.0
ΔTm (°C)

Figure 5: ΔT_m vs K_i fold-change.

- AB-343 maintains its favorable profile compared to nirmatrelvir and ensitrelvir against Mpro variants Y54A, F140A, S144A, L167F, and H172Y.
- For E166V variants, ΔT_m in presence of nirmatrelvir or AB-343 was minimal, consistent with resistance to these inhibitors in the biochemical assay.

CONCLUSIONS

- AB-343 has been identified as a novel inhibitor of SARS-CoV-2 Mpro with excellent pan-coronavirus activity (K_i 3 – 45 nM).
- Binding of AB-343 increases the T_m of Mpro by 23.2 °C, and dissociation is slow as measured by SPR.
- AB-343 potently inhibits Mpro from prevalent SARS-CoV-2 lineages, including Omicron (K_i 2 to 7 nM).
- AB-343 exhibits a favorable resistance profile against Mpro variants reported to confer resistance to other Mpro inhibitors in cell culture. AB-343 K_i fold changes against Y54A, F140A, S144A, L167F, and H172Y were modest (<6-fold), compared to nirmatrelvir (26 to 124-fold) and ensitrelvir (26 to 177-fold). Changes in ΔT_m of these variants are also less for AB-343 than for nirmatrelvir and ensitrelvir.
- Variants conferring resistance to Mpro inhibitors in general have reduced catalytic efficiency and lower thermal stability.
- AB-343 potently inhibits SARS-CoV-2 viral replication (EC_{50} = 18 nM) and exhibits a high selectivity index over human proteases and other viral proteases.[7]

REFERENCES

1. Collins, F. et al. (2023) The NIH-led research response to COVID-19. *Science* **379:** 441-444
2. Kuhlman, C. et al. (2022) Breakthrough infections with SARS-CoV-2 omicron despite mRNA vaccine booster dose. *Lancet* **399:** 625-626.
3. Shaffer, L. (2022) Lots of long COVID treatment leads, but few are proven. *Proc. Nat. Acad. Sci.* **119:** e2213524119
4. GISAID: https://gisaid.org/
5. Fact Sheet for Healthcare Providers: Emergency Use Authorization for Paxlovid™: https://www.fda.gov/media/155050/download
6. Jochmans, D. et al. (2023) The substitutions L50F, E166A, and L167F in SARS-CoV-2 3CLpro are selected by a protease inhibitor *in vitro* and confer resistance to nirmatrelvir. *mBio* **14**(1):e0281522
7. Mani, N. et al. *In Vitro* Antiviral Profile of AB-343, a Novel, Oral, Potent SARS-CoV-2 Mpro Inhibitor with Pan-coronavirus Activity. Poster presented at 36th International Conference on Antiviral Research; March 13-17, 2023; Lyon, France.

CONTACT INFORMATION AND DISCLOSURES

Kayleigh McGovern-Gooch, PhD.

Email: kmcgoverngooch@arbutusbio.com

Authors affiliated with Arbutus Biopharma are employees and may own company stock

www.arbutusbio.com