In Vitro Antiviral Profile of AB-343, a Novel, Oral, Potent SARS-CoV-2 Mpro Inhibitor with Pan-coronavirus Activity

Nagraj Mani*[1], Dimitar Gotchev[1], Kayleigh R. McGovern-Gooch[1], Andrzej Ardzinski[1], Andrea Cuconati[1], Rose Kowalski[1], Muhammad Sheraz[1], Holly M. Micolochick Steuer[1], Breanna Tercero[1], Xiaohe Wang[1], Adam Wasserman[1], Klaus Maskos[2], Chia-yi Chen[2], Konstanze von König[2], Andreas Schmitt[2], Archna Prasad[2], Michael Blaesse[2], Stefan Steinbacher[2], Debora L. Konz Makino[2], Kristi Yi Fan[1], Steven G. Kultgen[1], Aaron Lindstrom[1], Duyan Nguyen[1], Marvin Vega[1], Xu Wang[1], Amanda Pohl[1], Steven Ring[1], Fran Xu[1], Joseph Lucak[1], Troy O. Harasym[1], Andrew G. Cole[1], Angela M. Lam[1], and Michael J. Sofia[1]

[1]Arbutus Biopharma, Inc., Warminster, PA, USA and [2]Proteros Biostructures GmbH, D-82152 Martinsried, Germany.

13-17 MARCH 2023 ICAR2023 36TH International Conference on Antiviral Research (ICAR)

#532

Arbutus BIOPHARMA

INTRODUCTION

- The COVID-19 pandemic has resulted in significant mortality and morbidity: >700 million infected globally since Dec 2019 resulting in ~7 million deaths[1,2]
- Limited durability of vaccines require multiple, booster injections and the effectiveness of vaccines is continuously challenged by emerging new spike variants[3]
- Some individuals who have been infected with SARS-CoV-2 can experience long-term effects from their infection, known as post-COVID conditions (PCC) or long COVID[4]
- Current therapies either have suboptimal efficacy, inconvenient dosing route/regimen or require co-dosing with ritonavir that limits their broad application: creates the urgency and opportunity for developing more efficacious antivirals[5]
- The SARS-CoV-2 main protease (Mpro) is a cysteine protease responsible for the major polyprotein processing events and is essential for viral replication (Figure 1)
- Mpro is a clinically-validated target: due to the high degree of target site conservation across human coronaviruses, Mpro inhibitors have the potential to treat related human coronaviruses (SARS-CoV, MERS, OC43, HKU1, NL63, and 229E)[6]

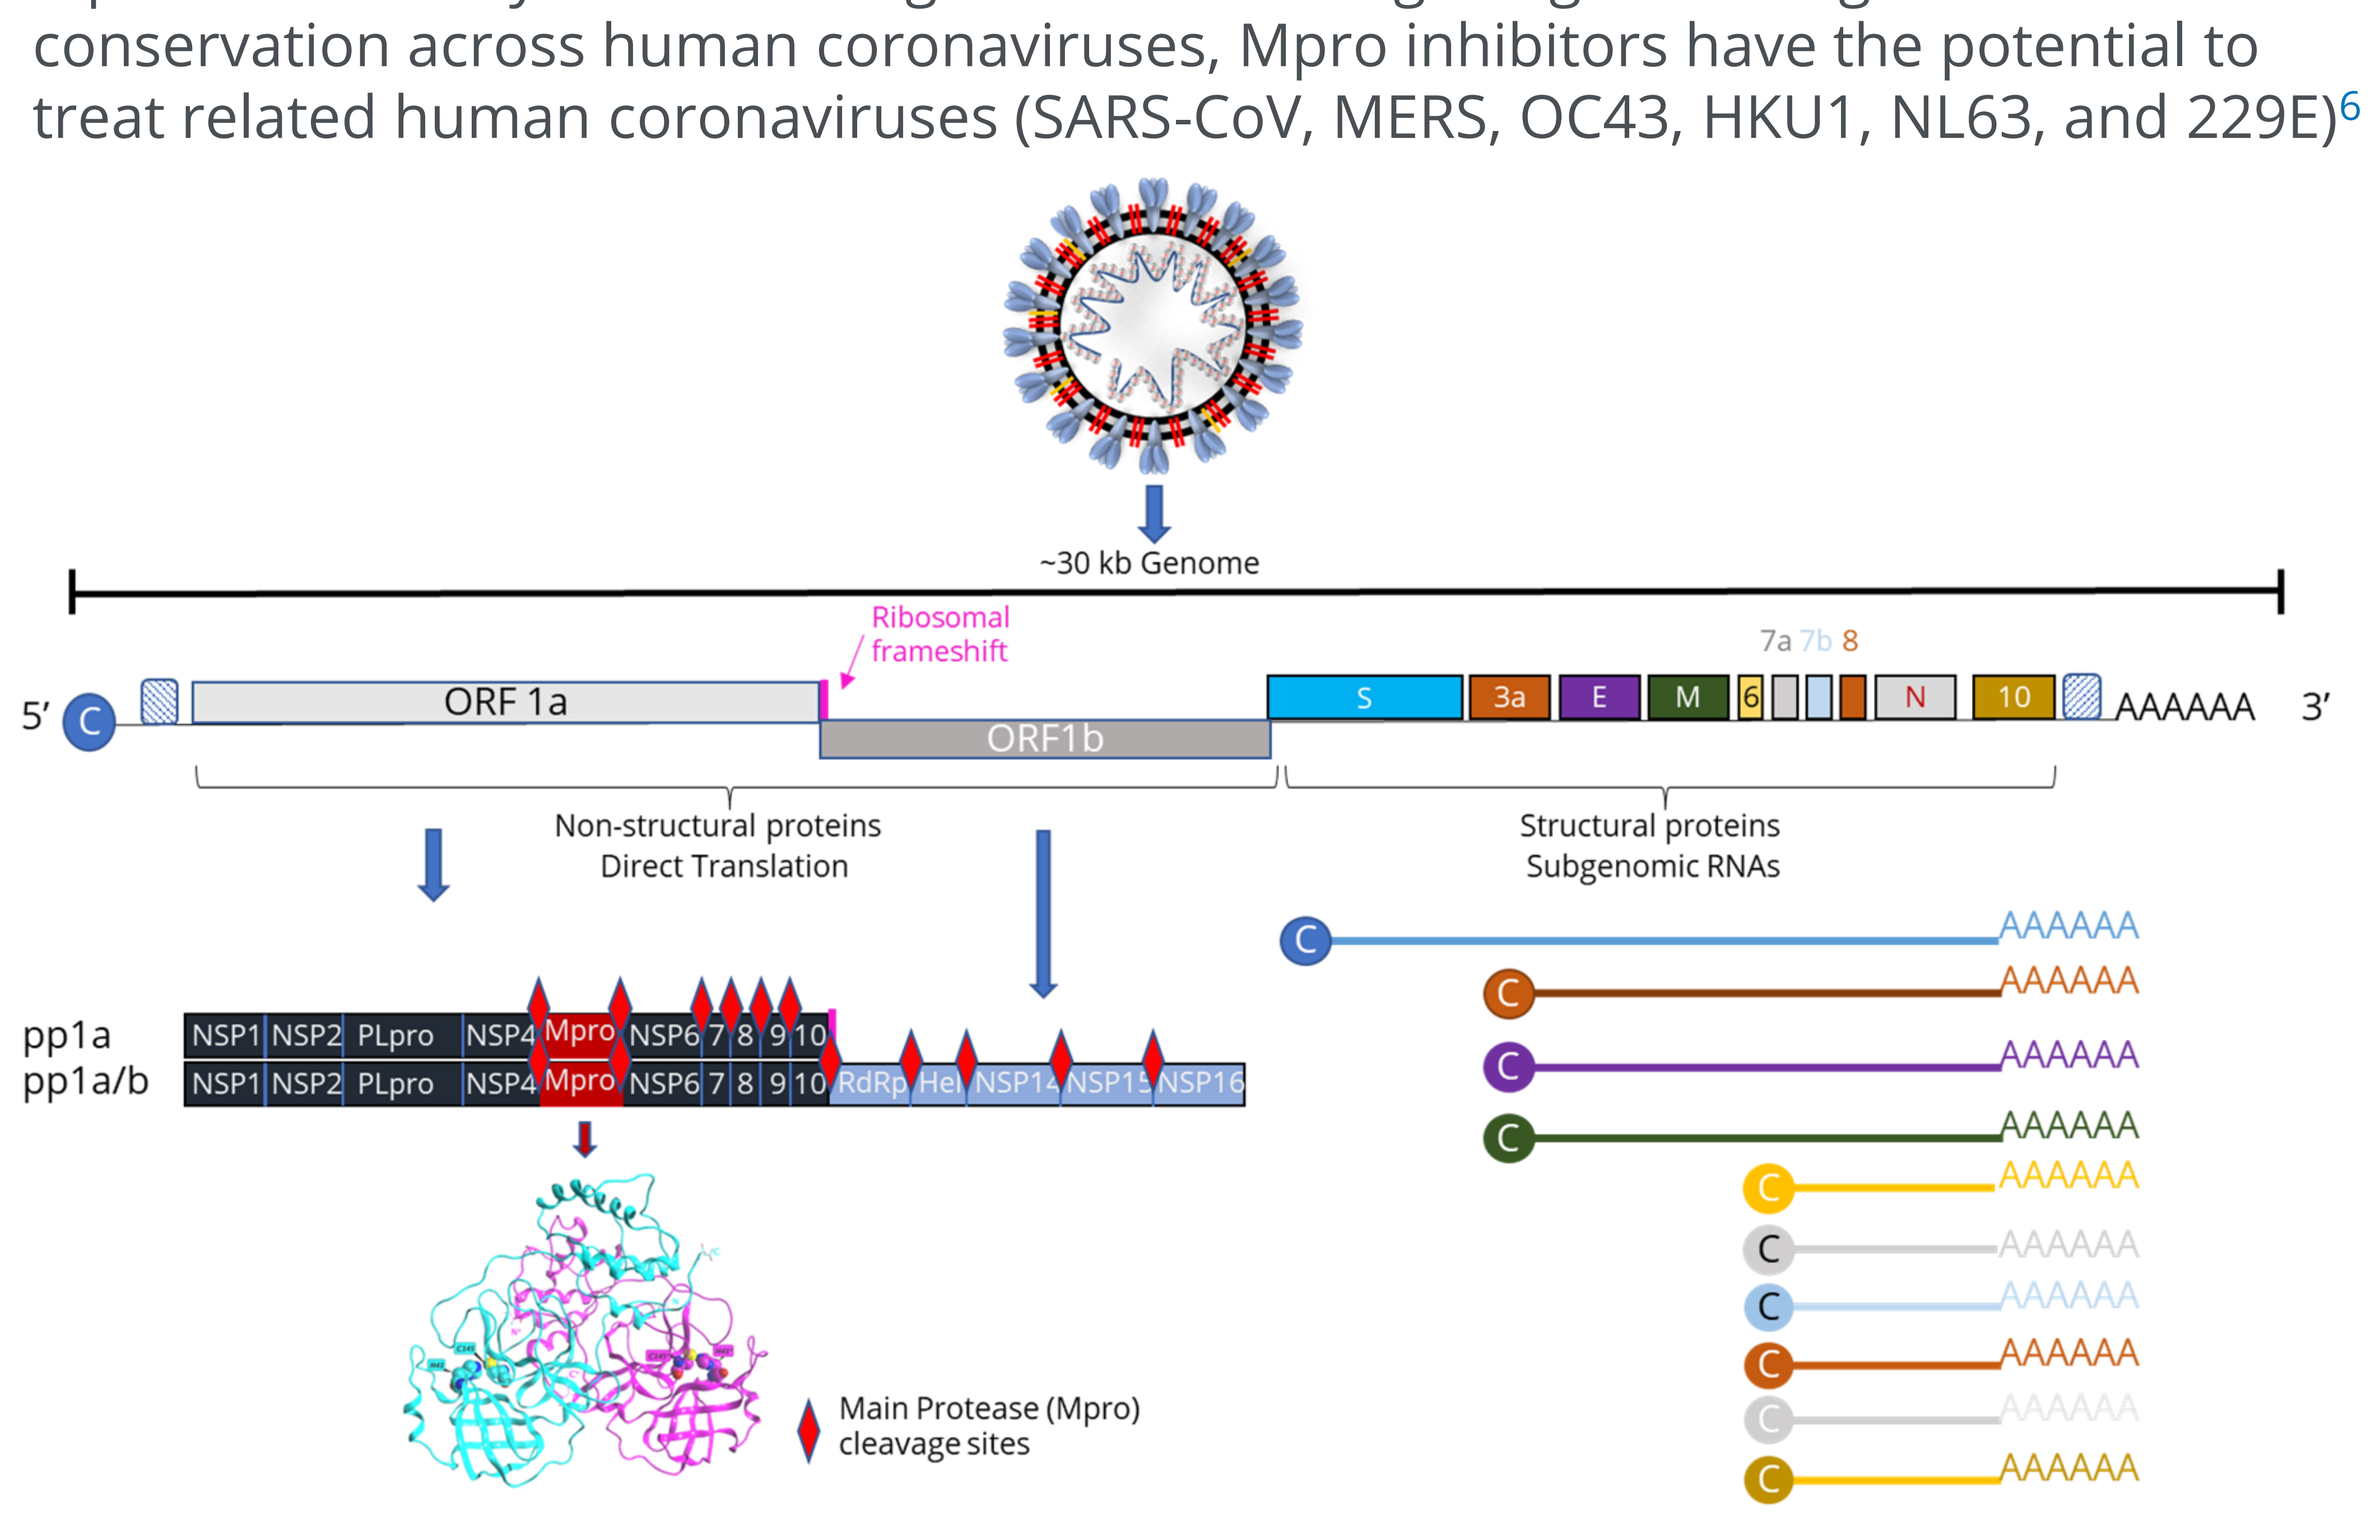
~30 kb Genome
Ribosomal frameshift
5′
ORF 1a
ORF1b
S 3a E M 6 7a 7b 8 N 10
AAAAAA 3′
Non-structural proteins
Direct Translation
Structural proteins
Subgenomic RNAs
pp1a
pp1a/b
NSP1 NSP2 PLpro NSP4 Mpro NSP6 7 8 9 10
RdRp Hel NSP14 NSP15 NSP16
Main Protease (Mpro) cleavage sites

Figure 1. Early replication events (genome expression and polyprotein processing) in the life cycle of SARS-CoV-2 and role of its main protease (Mpro)

METHODS

Enzyme K_i and catalytic efficiency measurements: Recombinant Mpro enzymes were expressed and purified from *E. coli*. Reactions consisted of Mpro, fluorogenic substrate, and Mpro inhibitors. Substrate cleavage was monitored over time and initial slopes were used to calculate fractional velocity to determine K_i values using the Morrison equation (GraphPad Prism). Catalytic efficiency was determined by titrating both substrate and enzyme, and initial slopes were calculated at each substrate concentration to determine K_M and K_{cat} using the Michaelis-Menten equation (GraphPad Prism).

Cathepsin A/B/L IC_{50} assays: Enzymes were preincubated with AB-343 for 30 min at room temperature (RT) and fluorescent peptide substrates were added and incubated for 60 min at RT. The fluorescent intensity signal was read at 360/500 nm, normalized to high and low signal controls to calculate % inhibition and plotted against AB-343 concentration to determine IC_{50} using a 4-parameter logistic equation using XLfit (IDBS, Boston).

Surface Plasmon Resonance (SPR): SPR measurements were performed using a Biacore system. Biotinylated Mpro was immobilized on SA chip (Series S) to reach around 1000 RU. 20 mM HEPES (pH7.5), 150 mM NaCl, 1 mM EDTA, 1 mM DTT, 0.05% T20 was applied as assay buffer for immobilization and analysis (plus 1% DMSO). The binding between testing compounds and Mpro were analyzed with SCK mode (single cycle kinetics) and kinetic parameters (k_{on}, k_{off} and K_D) were obtained using Biacore Insight Evaluation Software.

SARS-CoV-2 Infection assays: Antiviral activity against SARS-CoV-2 in infected VeroE6 cells (cytopathic effect (CPE) assay; in presence of a Pgp inhibitor) were performed by Imquest Biosciences (Frederick, MD). Activity against SARS-CoV-2 in infected A549_hACE2 cells with luciferase readout was conducted by P.Y.Shi lab (UTMB).

OC43 Infection assays: Antiviral activity against OC43 was measured using a CPE assay in infected MRC-5 cells and in infected Huh7 cells using a bDNA assay.

Mpro database sequence analyses: The prevalence of Mpro variants was determined by mining the SARS-CoV-2 sequences in the GISAID database (https://gisaid.org/).

Human protease panel assays: Human protease panel AB-343 IC_{50} values were determined by Reaction Biology (Malvern, PA).

Virus panel selectivity assays: Activity of AB-343 against a virus panel involving cell-based infectious systems were conducted by Imquest Biosciences (Frederick, MD) and/or their subcontractor(s).

RESULTS

AB-343 targets SARS-CoV-2 Mpro and inhibits viral replication:

- AB-343 is a potent inhibitor of SARS-CoV-2 Mpro enzyme activity
- In an SPR assay, binding of AB-343 to SARS-CoV-2 Mpro resulted in longer enzyme:ligand residence times indicative of excellent target engagement
- Consistent with enzyme inhibition, AB-343 inhibited SARS-CoV-2 replication in cell-based infectious virus assays

Table 1. *In vitro* activities of AB-343 against SARS-CoV-2 in enzyme and cell-based assays

SARS-CoV-2 Mpro Assays	Nirmatrelvir	Ensitrelvir	AB-343
Mechanism (competitive binding to catalytic site):	Covalent	Non-covalent	Covalent
SARS-CoV-2 Mpro: Residence Time by SPR (min)	21.5	1.1	>99
SARS-CoV-2 Mpro: K_i (μM)	0.001 ± 0.0003	0.001 ± 0.0005	0.003 ± 0.0008
SARS-CoV-2_VeroE6_CPE (+PgpI): EC_{50} \| CC_{50} (μM)	0.029 ± 0.027\| >3	0.087 ± 0.091 \| >3	0.020 ± 0.005\| >30
SARS-CoV-2_A549/hACE2_Luc: EC_{50} \| CC_{50} (μM)	0.034 ± 0.017\| >50	nd	0.018 ± 0.009\|>10

Notes: PgpI = P-glycoprotein inhibitor CP-100356 (2 μM); Nirmatrelvir is PF-07321332, the Mpro inhibitor in Paxlovid™ (Pfizer); Ensitrelvir is S-217622 the active ingredient in Xocova® (Shionogi). Enzyme and cell-based potency determination values are average and SD of at least 3 independent determinations. K_i, EC_{50} and CC_{50} data on nirmatrelvir and ensitrelvir shown were generated by Arbutus. nd – not determined.

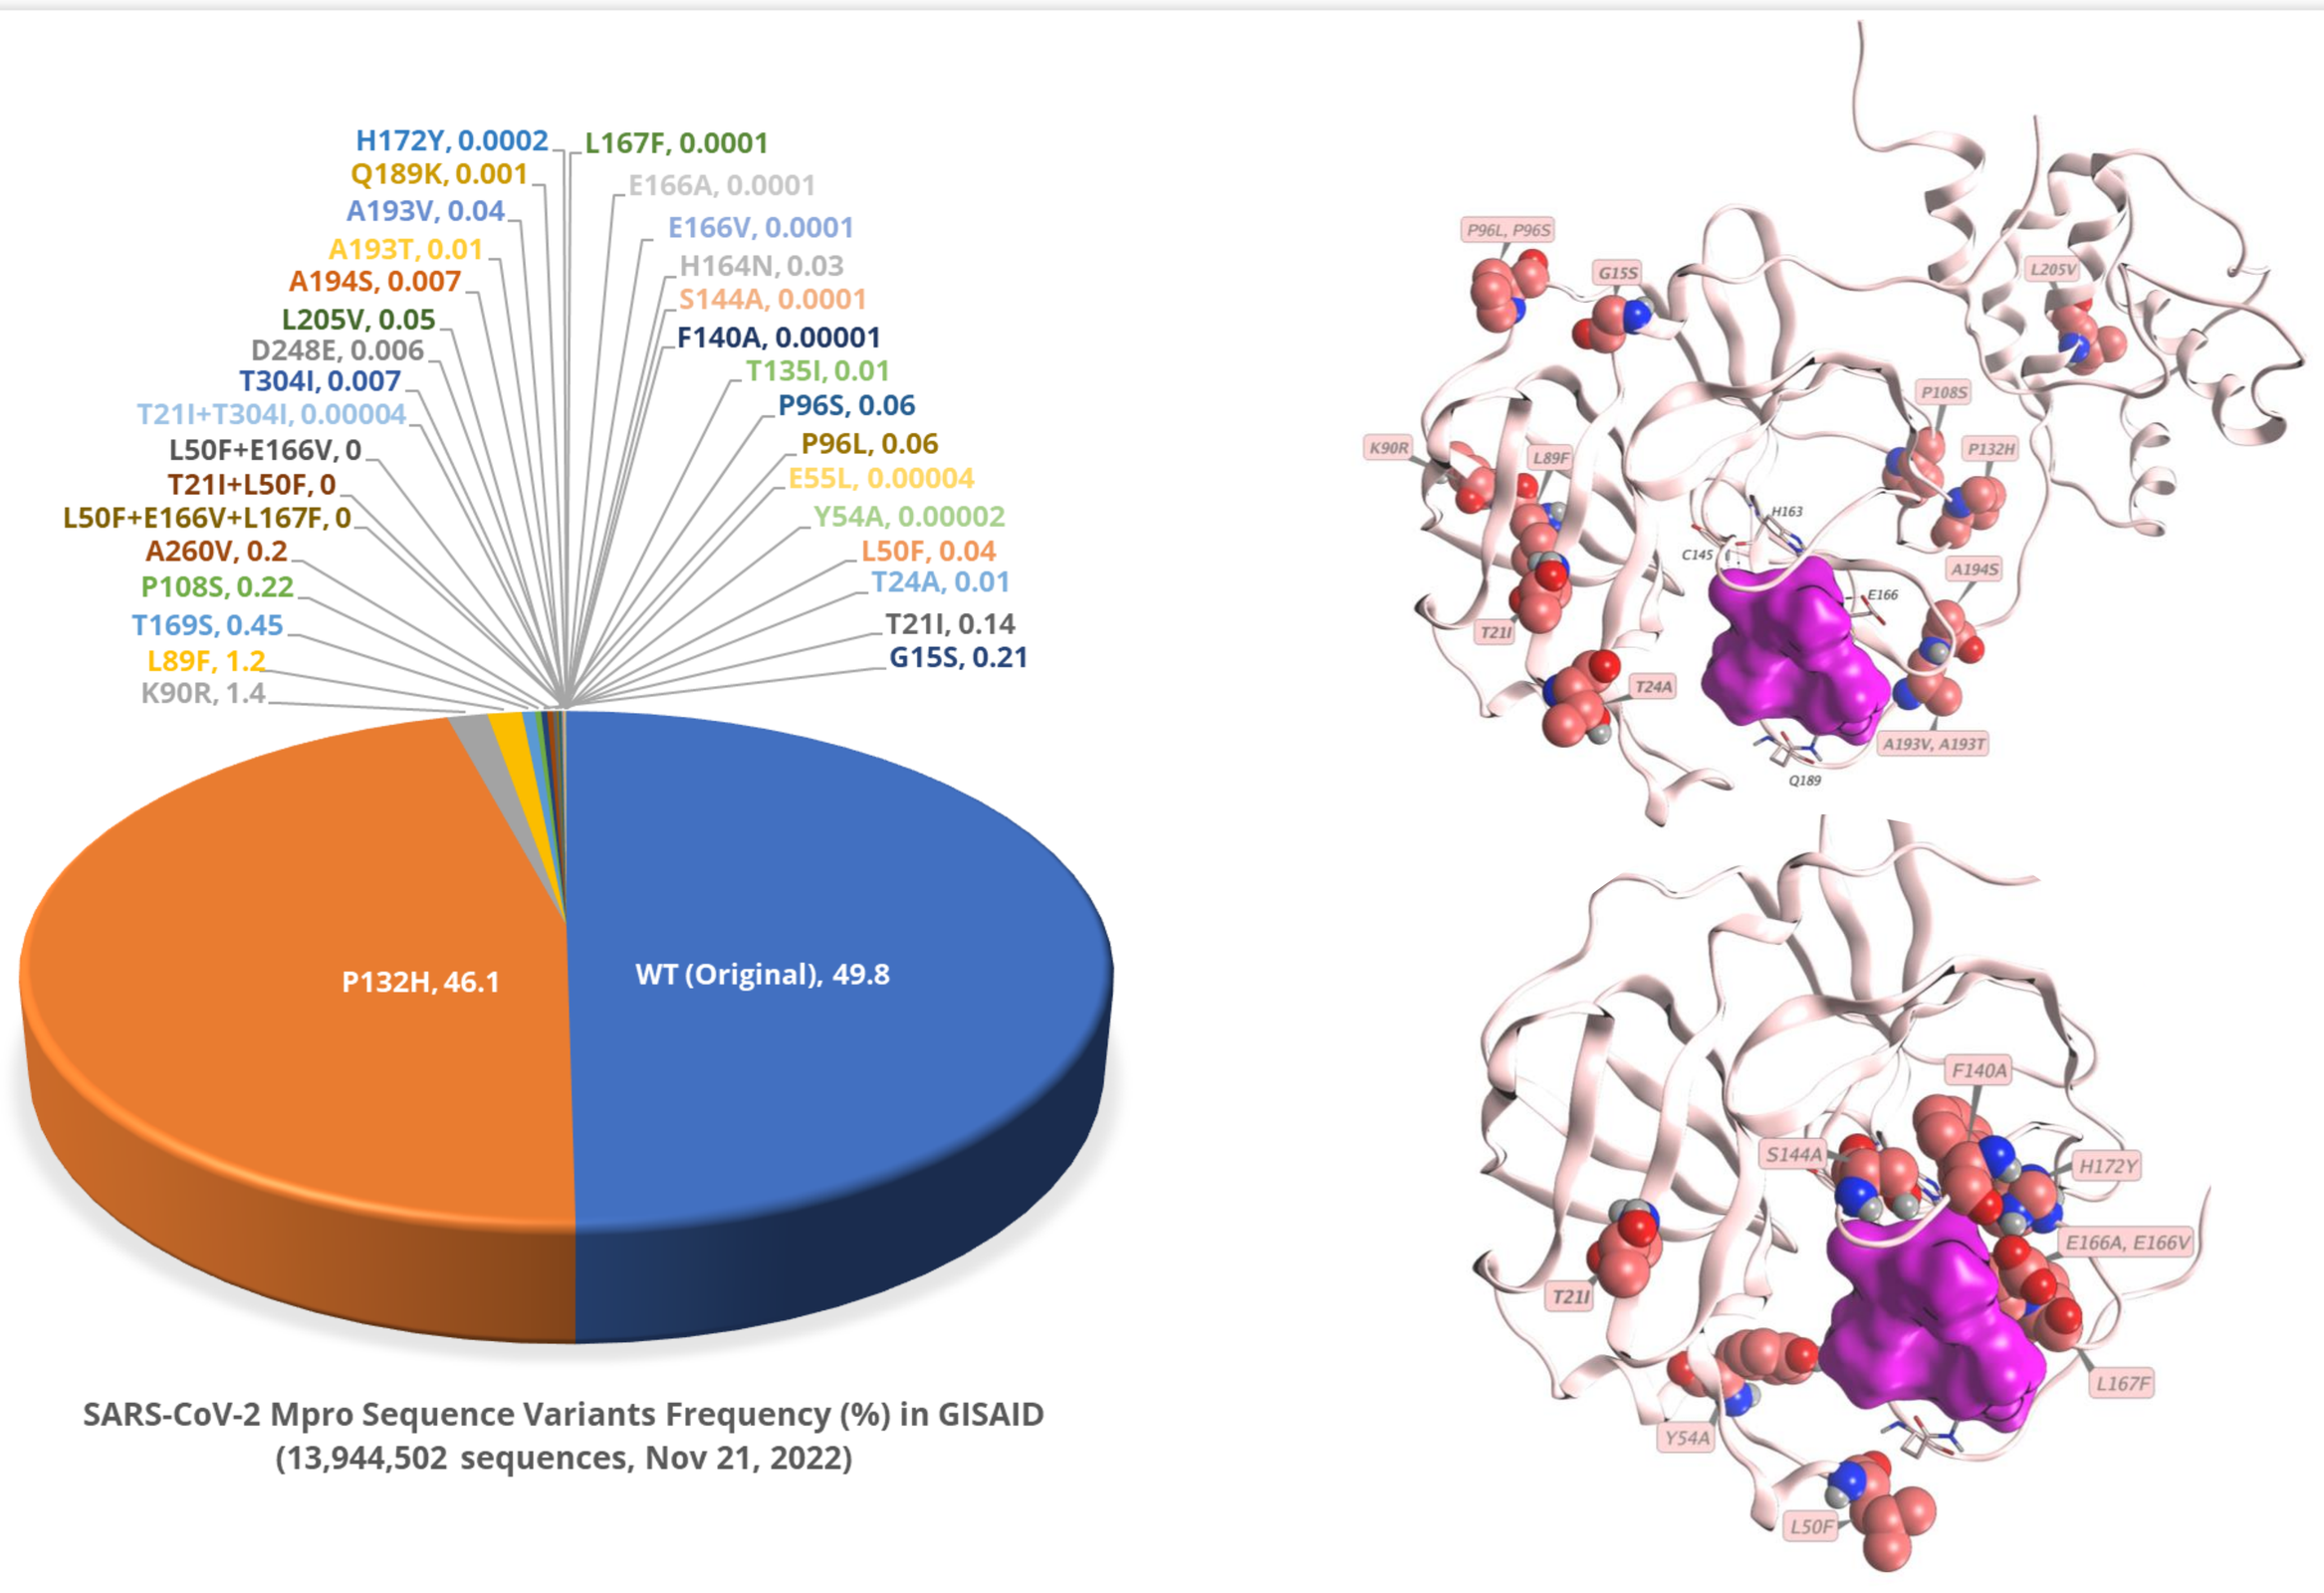
H172Y, 0.0002
L167F, 0.0001
Q189K, 0.001
E166A, 0.0001
A193V, 0.04
E166V, 0.0001
A193T, 0.01
H164N, 0.03
A194S, 0.007
S144A, 0.0001
L205V, 0.05
F140A, 0.00001
D248E, 0.006
T135I, 0.01
T304I, 0.007
P96S, 0.06
T21I+T304I, 0.00004
P96L, 0.06
L50F+E166V, 0
E55L, 0.00004
T21I+L50F, 0
Y54A, 0.00002
L50F+E166V+L167F, 0
L50F, 0.04
A260V, 0.2
T24A, 0.01
P108S, 0.22
T21I, 0.14
T169S, 0.45
G15S, 0.21
L89F, 1.2
K90R, 1.4
P132H, 46.1
WT (Original), 49.8
SARS-CoV-2 Mpro Sequence Variants Frequency (%) in GISAID
(13,944,502 sequences, Nov 21, 2022)

Figure 2. Natural prevalence of SARS-CoV-2 Mpro variants in GISAID and their locations in relation to AB-343 bound to the catalytic site

AB-343 inhibits Mpro variants from prevalent SARS-CoV-2 lineages:

- Naturally prevalent SARS-CoV-2 Mpro enzyme variants were cloned, expressed and purified from *E. coli* and tested for inhibition by AB-343
- Data suggests that these variants show uniform susceptibility to AB-343

Table 3: Activity of AB-343 against Mpro enzyme sequence variants *in vitro*

SARS-CoV-2 Mpro Enzyme Variant	SARS-CoV-2 Lineage	Frequency (%) in GISAID database**	Nirmatrelvir K_i Fold Increase	Ensitrelvir K_i Fold Increase	AB-343 K_i Fold Increase
WT (original)	Original	49.8	1.0	1.0	1.0
G15S	C.37 Lambda	0.21	1.5	1.2	1.2
T21I	B.1.1.318	0.14	1.0	0.6	0.7
T24A	B.1.524	0.01	1.0	1.5	0.8
L89F	B.1.2	1.20	1.0	2.0	0.6
K90R	B.1.351 Beta	1.40	1.0	0.9	0.9
P96L	B.1.1.419	0.06	1.4	1.0	0.9
P96S	B.1.1.389	0.06	1.2	0.7	1.0
P108S	N. 10	0.22	1.2	1.0	0.7
P132H	Omicron	46.1	1.3	1.3	0.9
A193V	B.1.351.2 Beta	0.04	1.0	0.7	0.8
A193T	-	0.01	1.0	0.8	0.9
A194S	B.1.617.3	0.01	3.5	2.2	2.4
L205V	P.2 Zeta	0.05	0.9	0.8	0.7

Notes: **Natural prevalence in 13,944,502 GISAID sequences; Nov 21, 2022; Data on nirmatrelvir (PF-07321322) and ensitrelvir (S-217622) shown were generated by Arbutus.

AB-343 shows a differentiating *in vitro* resistance profile vs nirmatrelvir and ensitrelvir:

- AB-343 was also tested in enzyme assays against SARS-CoV-2 Mpro variants isolated in cell culture and known to reduce the susceptibility of other Mpro inhibitors
- AB-343 showed lower fold-increases in K_i for SARS-CoV-2 Mpro variants Y54A, F140A, S144A, L167F, and H172Y in comparison to nirmatrelvir and ensitrelvir
- In comparison, cross-resistance was observed amongst nirmatrelvir, ensitrelvir, and AB-343 for SARS-CoV-2 Mpro variants H164N and E166A/V; however, these variants demonstrated low catalytic efficiency (Figure 3).

Table 4. Activity of AB-343 against Mpro variants known to reduce *in vitro* susceptibility to SARS-CoV-2 Mpro inhibitors

Mpro Enzyme Variant	Frequency (%) in GISAID database**	Nirmatrelvir K_i Fold Increase	Ensitrelvir K_i Fold Increase	AB-343 K_i Fold Increase
Y54A	0.00002	28.8	89.2	2.2
F140A	-	41.7	68.1	3.6
S144A	0.0001	26.0	64.7	2.3
H164N	0.034	5.8	11.6	5.0
E166A	0.0001	90.9	104.8	40.7
E166V	0.0001	>14203	600.2	>3597
L167F	0.0001	27.7	25.7	2.6
H172Y	0.0002	123.8	176.6	5.4
L50F	0.04	1.0	0.3	0.7
L50F + E166V	-	>14203	204.1	2333
L50F + E166V + L167F	-	>14203	1366	>3597

Notes: **Natural prevalence in 13,944,502 GISAID sequences; Nov 21, 2022; Fold change in K_i's were calculated as a ratio of mutant enzyme average K_i divided by the WT average Ki. Data on nirmatrelvir (PF-07321322) and ensitrelvir (S-217622) shown were generated by Arbutus.

Table 5. AB-343 is highly selective for SARS-CoV-2 in comparison to a panel of human proteases and viruses representing different virus families

Protease Assay	AB-343 IC_{50} (μM)
Cathepsin A (Serine)	>44
Cathepsin B (Cysteine)	>44
Cathepsin D (Aspartyl)	>30
Cathepsin K* (Cysteine)	~30
Cathepsin L (Cysteine)	>44
Cathepsin S (Cysteine)	>30
Calpain 1 (Cysteine)	>30
Chymotrypsin (Serine)	>30
Caspase 2 (Cysteine)	33.7
Caspase 3 (Cysteine)	>30
Elastase (Serine)	>30
Furin (Serine)	>30
Thrombin a (Serine)	>30
TMPRSS2 (Serine)	>30
HIV (Aspartyl)	>30
SARS-CoV-2 PLpro	>50
SARS-CoV-2 Mpro	0.008

Virus/Host	Family	Genome	AB-343 EC_{50} (μM)	AB-343 CC_{50} (μM)
HCV/Huh7	Flaviviridae	(+) ssRNA	1.99	>30
WNV/Vero76	Flaviviridae	(+) ssRNA	>30	>30
RSV/HEp2	Paramyxoviridae	(-) ssRNA (nonseg.)	>30	>30
FluA/MDCK	Orthomyxoviridae	(-) ssRNA (seg.)	>30	>30
HIV/CEMSS	Retroviridae	ssRNA to DNA	>30	>30
HSV1/Vero	Herpesviridae	dsDNA	>30	>30
HSV2/Vero	Herpesviridae	dsDNA	>30	>30
HCMV/MRC5	Herpesviridae	dsDNA	>30	>30
DENV/BHK21	Flaviviridae	(+) ssRNA	>30	>30
HRV/H1-HeLa	Picornaviridae	(+) ssRNA	>30	>30

HCV = hepatitis C virus; WNV = west nile virus; RSV = respiratory syncytial virus; HIV = human immunodeficiency virus; HSV-1/2 = herpes simplex virus-1/2; hCMV = human cytomegalovirus; DENV = dengue virus; HRV = human rhinovirus

- AB-343 shows high degree of selectivity *vs* a human protease panel
- AB-343 showed no inhibition of HIV-1 protease and SARS-CoV-2 PLpro
- AB-343 did not show inhibition against a diverse panel of viruses representing different families except HCV (EC_{50} of ~2 μM)

AB-343 shows potent pan-coronavirus activity:

- AB-343 was evaluated for activity against Mpro enzymes derived from human coronaviruses SARS-CoV, MERS, HKU1, NL63, 229E, and OC43
- AB-343 showed pan-coronavirus activity with K_i's in the range of 5 – 45 nM
- In a cell-based assay of OC43, the potent inhibition of OC43 Mpro enzyme translated into potent inhibition of viral replication

Table 2. Pan-coronavirus profile of AB-343 in enzyme and cell-based assays

Mpro Enzyme Assay	Nirmatrelvir K_i (μM)	Ensitrelvir K_i (μM)	AB-343 K_i (μM)
SARS-CoV	0.002 ± 0.0003	0.001 ± 0.0004	0.005 ± 0.003
MERS	0.035 ± 0.012	0.292 ± 0.191	0.045 ± 0.006
HKU1	0.005 ± 0.002	0.003 ± 0.001	0.006 ± 0.002
NL63	0.142 ± 0.064	>5	0.039 ± 0.005
229E	0.055 ± 0.020	>5	0.036 ± 0.014
OC43	0.011 ± 0.005	0.012 ± 0.004	0.019 ± 0.004
Cell Based Assay	EC_{50} \| EC_{90} \| CC_{50} (μM)	EC_{50} \| EC_{90} \| CC_{50} (μM)	EC_{50} \| EC_{90} \| CC_{50} (μM)
OC43_MRC5_CPE	0.128 \| 0.290 \| >50	0.244 \| 0.341 \| >50	0.203 \| 0.251 \| >50
OC43_Huh7_bDNA	0.110 \| 0.151 \| >25	0.380 \| 0.435 \| >25	0.102 \| 0.120 \| >25

Notes: nirmatrelvir is PF-07321332, the Mpro inhibitor in PAXLOVID™ (Pfizer); ensitrelvir is S-217622 the active ingredient in Xocova® (Shionogi); Values are averages of at least n ≥ 2.

Catalytic efficiency of SARS-CoV-2 Mpro shows inverse proportionality to K_i fold-change:

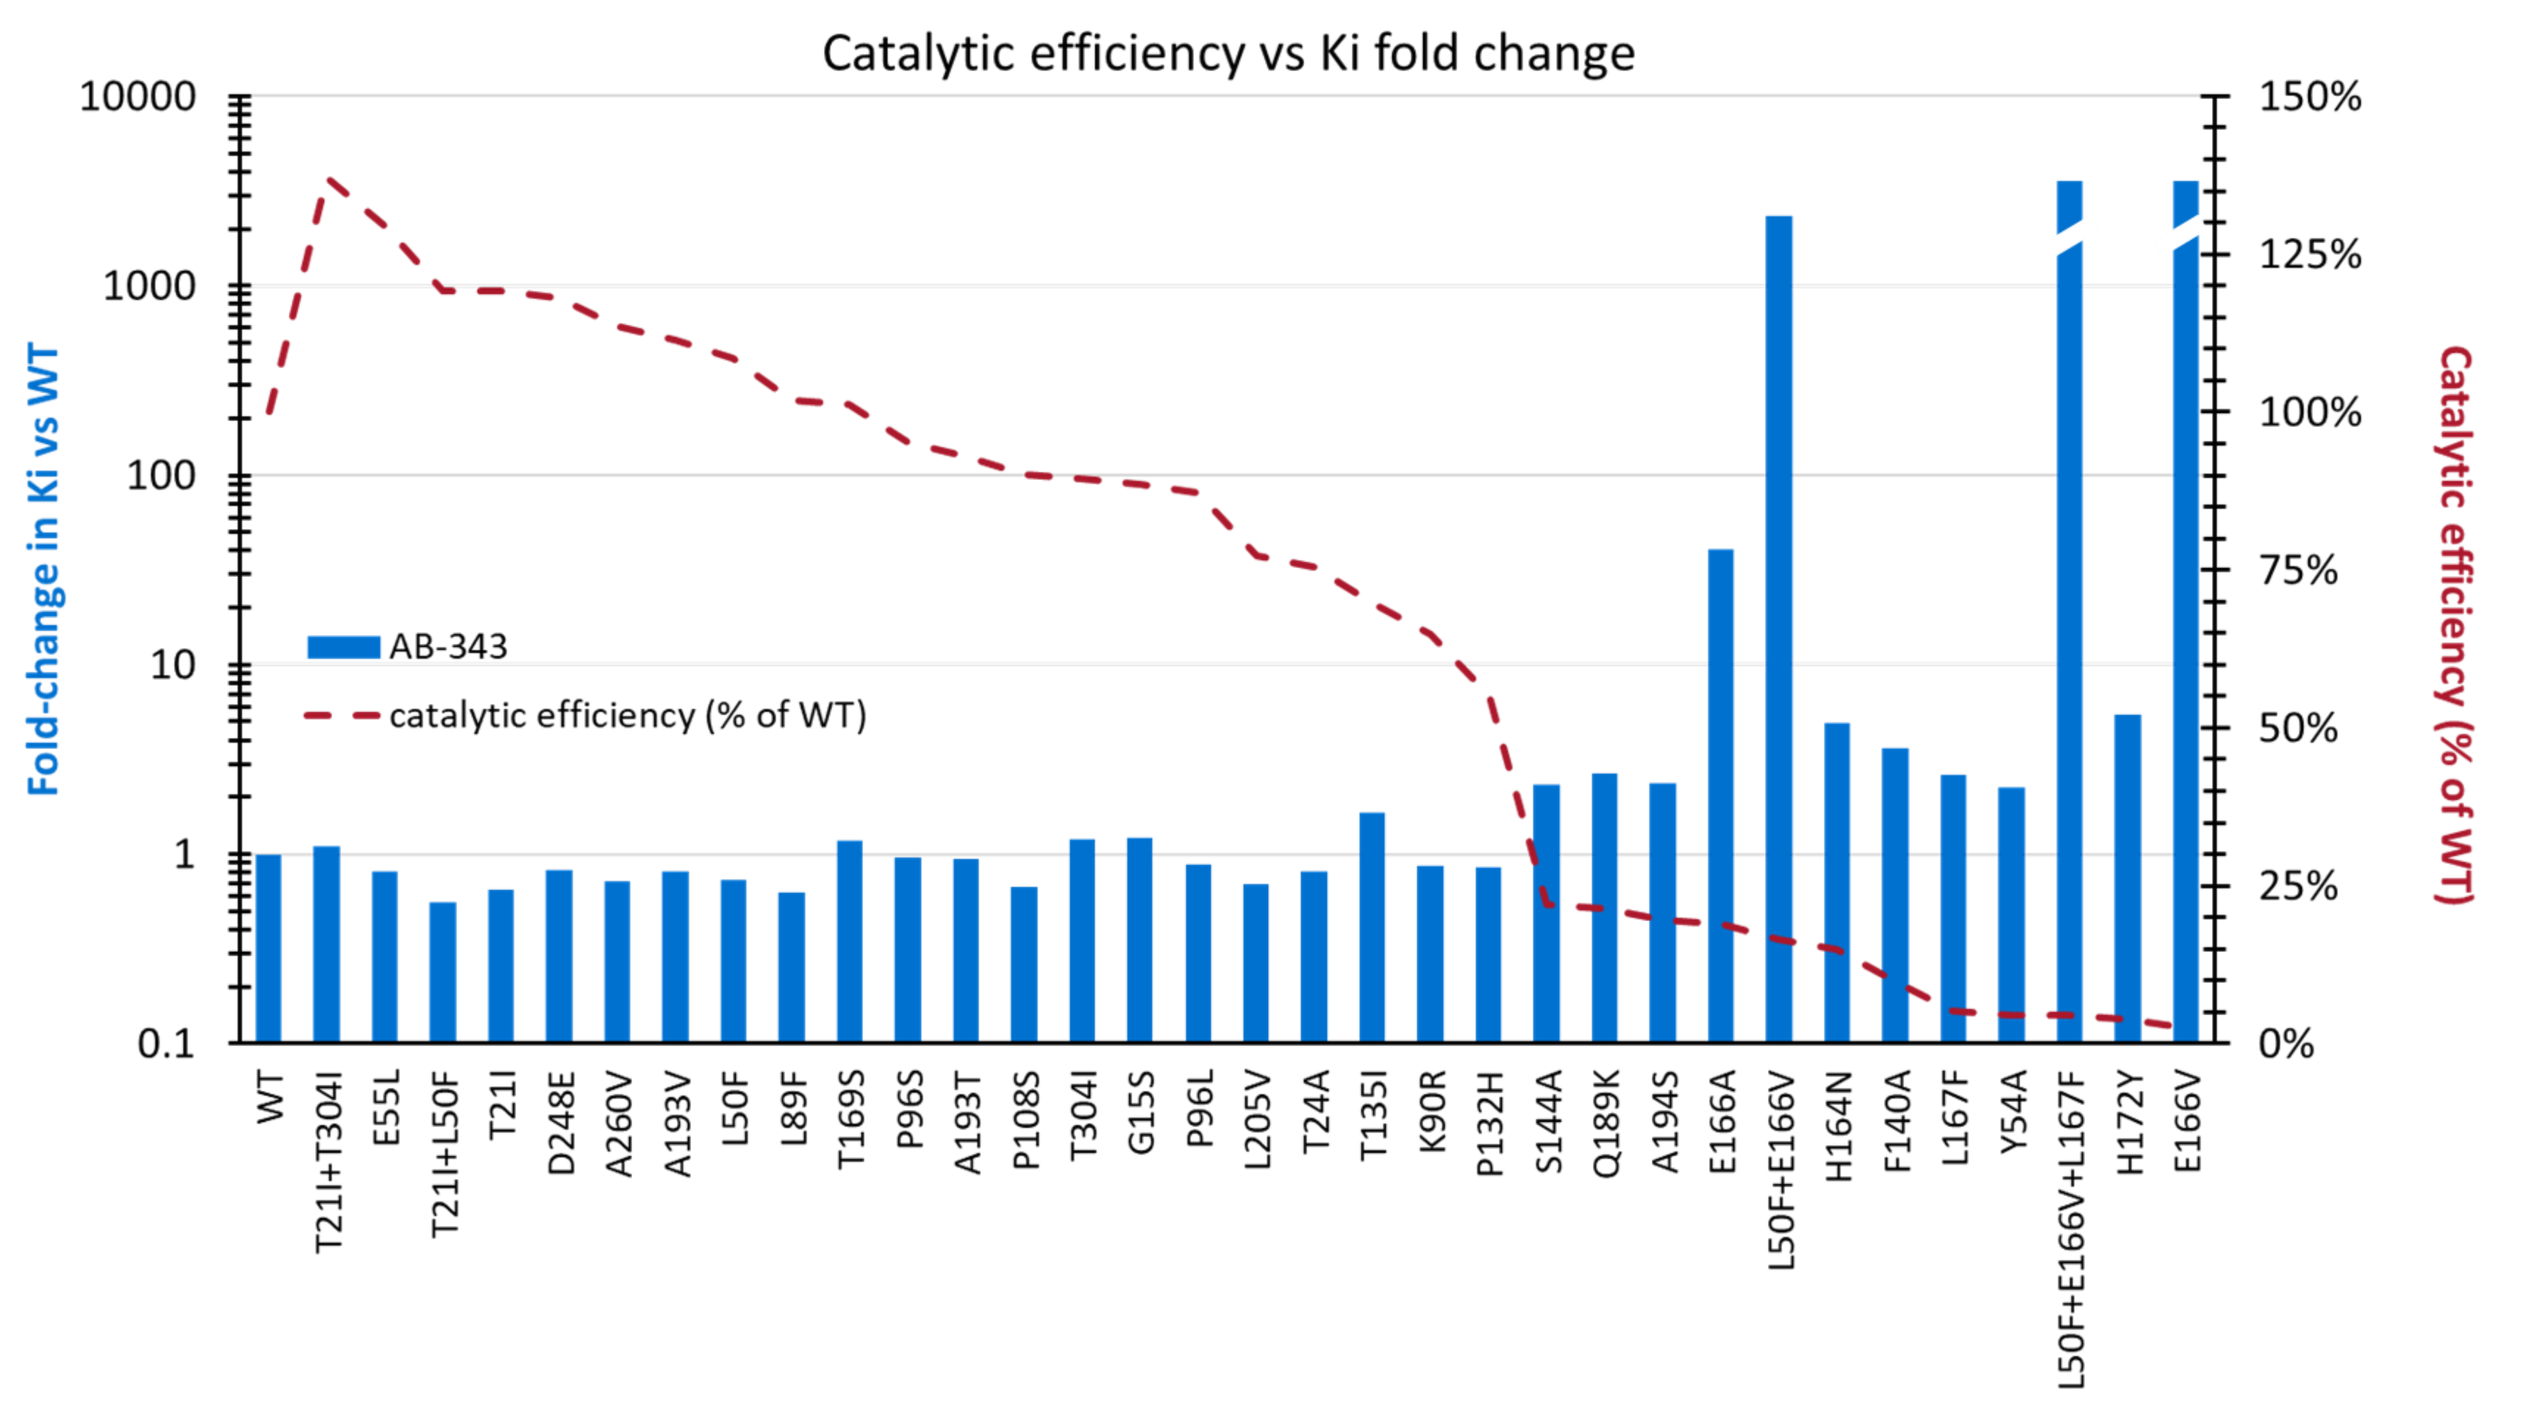
Catalytic efficiency vs Ki fold change
Fold-change in Ki vs WT
Catalytic efficiency (% of WT)
10000
1000
100
10
1
0.1
150%
125%
100%
75%
50%
25%
0%
AB-343
catalytic efficiency (% of WT)
WT
T21I+T304I
E55L
T21I+L50F
T21I
D248E
A260V
A193V
L50F
L89F
T169S
P96S
A193T
P108S
T304I
G15S
P96L
L205V
T24A
T135I
K90R
P132H
S144A
Q189K
A194S
E166A
L50F+E166V
H164N
F140A
L167F
Y54A
L50F+E166V+L167F
H172Y
E166V

Figure 3. Relationship between Mpro catalytic efficiency of variants *vs* AB-343 Mpro K_i

CONCLUSIONS

- AB-343 is a potent inhibitor of SARS-CoV-2 Mpro (K_i 0.003 μM) and efficiently inhibits SARS-CoV-2 replication *in vitro* (EC_{50} 0.018 μM).
- X-ray structural analysis of AB-343 bound to SARS-CoV-2 Mpro confirmed covalent association with the catalytic cysteine sidechain in addition to a network of electrostatic and steric interactions supporting the excellent *in vitro* potency profile.
- AB-343 also demonstrated pan-coronavirus inhibition with a K_i range of 0.005 to 0.045 μM against Mpro enzymes from multiple coronaviruses.
- AB-343 maintained potency against naturally prevalent SARS-CoV-2 Mpro enzyme variants including P132H (Omicron).
- Among Mpro variants known to confer *in vitro* resistance to other Mpro inhibitors, the K_i increases for AB-343 against SARS-CoV-2 Mpro Y54A, F140A, S144A, L167F, and H172Y were modest (2 to 5-fold) in comparison to nirmatrelvir (26 to 124-fold) and ensitrelvir (26 to177-fold). However, these Mpro variants were rare in the GISAID database and high-level *in vitro* resistance inversely correlated with the catalytic efficiency of SARS-CoV-2 Mpro.
- AB-343 showed little to no inhibition of an enzyme panel comprising human and viral proteases demonstrating its high selectivity for coronavirus Mpro.
- AB-343 also showed no significant replication inhibition of viruses from different families with EC_{50} and CC_{50} of >30 μM, except HCV (EC_{50} 2 μM).
- Based on its antiviral potency, selectivity, and favorable PK, AB-343 was selected for further development as a potential ritonavir-free oral treatment for COVID-19 and other coronaviruses.

REFERENCES

1. WHO Coronavirus Dashboard: https://covid19.who.int/
2. Collins F. *et al.* 2023. The NIH-led research response to COVID-19. *Science* **379:** 441-444
3. US Centers of Disease Control and Prevention: https://www.cdc.gov/coronavirus/2019-ncov/long-term-effects/index.html
4. Liu M. and Li Y. 2022: Advances in COVID-19 Vaccines and new coronavirus variants. *Front. Med*. **9:** 888631
5. Gandhi RT, Malani PN, del Rio C. 2022. COVID-19 Therapeutics for nonhospitalized patients. *JAMA*. **327(7):** 617–618
6. Cannalire R, *et al.* 2022. Targeting SARS-CoV-2 proteases and polymerase for COVID-19 treatment: state of the art and future opportunities. *J Med Chem.* 65(4):2716-2746. Epub 2020 Nov 13
7. Duveau, D.Y. and Thomas, C.J. 2022: The remarkable selectivity of nirmatrelvir. *ACS Pharmacol. Transl. Sci*. **5:** 445−447

CONTACT INFORMATION AND DISCLOSURES

Nagraj Mani, PhD.
Arbutus Biopharma, Inc.,
701 Veterans Circle,
Warminster, PA 18974
Email: nmani@arbutusbio.com

Authors affiliated with Arbutus Biopharma are employees and may own company stock

Acknowledgments (Proteros): Astrid Hampel, Anita Schiller, Daniela Gotz, Martin Augustin (Protein Sciences); Irina Trotsenko, Claudia Martinez de Baum (SPR); Marc Hellmann, Patience Sithole (IC_{50} assays); Shobana Murugan (X-ray crystallography)

WEBSITE:

www.arbutusbio.com